4SC-202 + Pembrolizumab – „Sensitivierung“ PD-1-refraktärer Melanome zur Erhöhung der Wirksamkeit von Checkpoint-Blockade bei Patienten mit fortgeschrittenem Melanom

4SC-202 + Pembrolizumab: “SENSITIZE” the tumor to overcome PD-1 refractoriness and increase efficacy of checkpoint inhibition in patients with advanced melanoma

Bartz R., Baumgartner R., Ammendola A., Hamm S., Hermann F.

4SC AG, Martinsried; Germany

P125

Abstract

Despite successes in the treatment of melanoma patients with checkpoint inhibitors (anti-PD1 antibodies), the majority of patients do not respond to checkpoint inhibition alone and a high unmet medical need remains for these patients. One promising approach is to increase the number of patients benefiting from checkpoint inhibition by enhancing the immunogenicity and alter the tumor microenvironment from a more immune-deserted to an immune-inflamed phenotype by means of combination therapy. Epigenetic modulation has been reported as one key determining factor in shaping the immune microenvironment and compounds altering these processes (e.g. histone deacetylases (HDAC) inhibitors) are particularly promising.
Here, we report results for 4SC-202, an orally available clinical stage HDAC inhibitor, and outline the further clinical development. 4SC-202 treatment led to an increase of MHC class II molecules and enhanced expression of inflammatory markers like IFN-γ and various chemokines in tumors. Furthermore, detailed analysis of the tumor microenvironment in tumor bearing animals revealed that 4SC-202 strongly altered the immune cell composition and particularly the number of cytotoxic T cells (CTL) was markedly increased. Importantly, subsequent combination treatment of 4SC-202 with checkpoint inhibitors in syngenic animal models showed a strong synergistic effect resulting in an increased tumor growth reduction.
For the further clinical development, start of a Phase Ib/II clinical study (‘SENSITIZE’) is planned by the end of this year. This study, conducted in Germany with up to 6 sites (~30 patients) will enroll patients with advanced cutaneous melanoma who are refractory/non-responding to treatment with anti-PD-1 antibodies. These patients clearly represent a population with a high unmet medical need and might be characterized by an unfavorable tumor immunology and microenvironment for immunotherapy in general and checkpoint inhibition in particular. We hypothesize that addition of 4SC-202 to anti-PD-1 antibody treatment may lead to increased immunogenicity of the tumor, an inflamed tumor microenvironment and ultimately to clinical benefit in anti-PD-1 refractory/non-responding advanced melanoma patients.

Treatment with 4SC-202 leads to increased infiltration of CD8+ T-cells into tumor tissue.

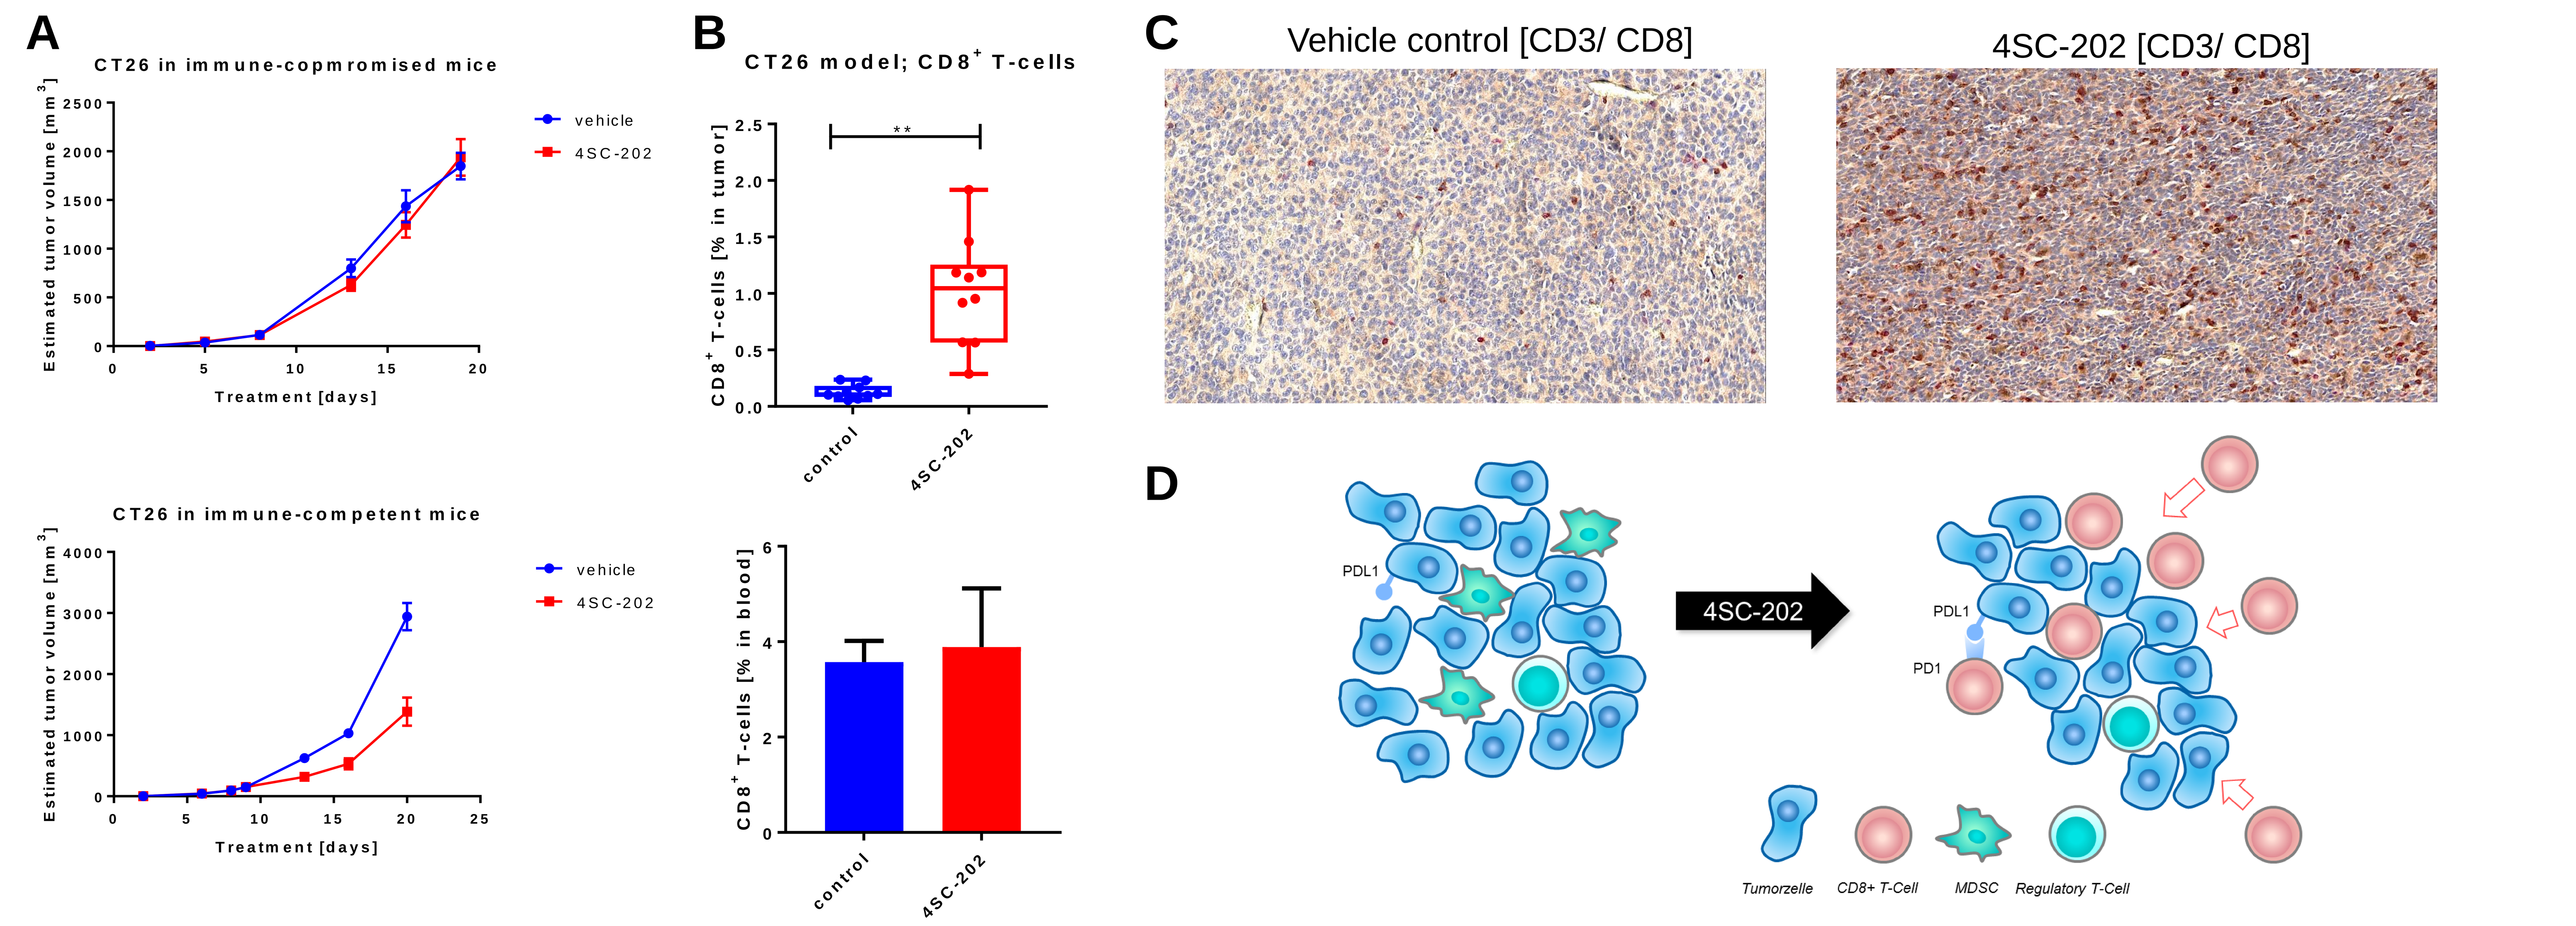

A
CT26 in immune-copmromised mice
Estimated tumor volume [mm3]
Treatment [days]
vehicle
4SC-202
CT26 in immune-competent mice
B
CT26 model; CD8+ T-cells
CD8+ T-cells [% in tumor]
**
CD8+ T-cells [% in blood]
control
4SC-202
C
Vehicle control [CD3/ CD8]
4SC-202 [CD3/ CD8]
D
PDL1
4SC-202
PD1
Tumorzelle
CD8+ T-Cell
MDSC
Regulatory T-Cell

Fig. 1: (A) Anti-tumor activity of 4SC-202 requires a functional immune system. **(B, C)** 4SC-202 enhances infiltration with cytotoxic CD8⁺ T-cells in TME of CT26 tumors, whereas the total levels of CD8⁺ T-cells remain unchanged. **(D)** Proposed mechanism of action: epigenetic modulation by 4SC-202 increases the recruitment of effector cells into the tumor rendering it more susceptible to treatment with checkpoint inhibitors.

Pre-clinical animal models

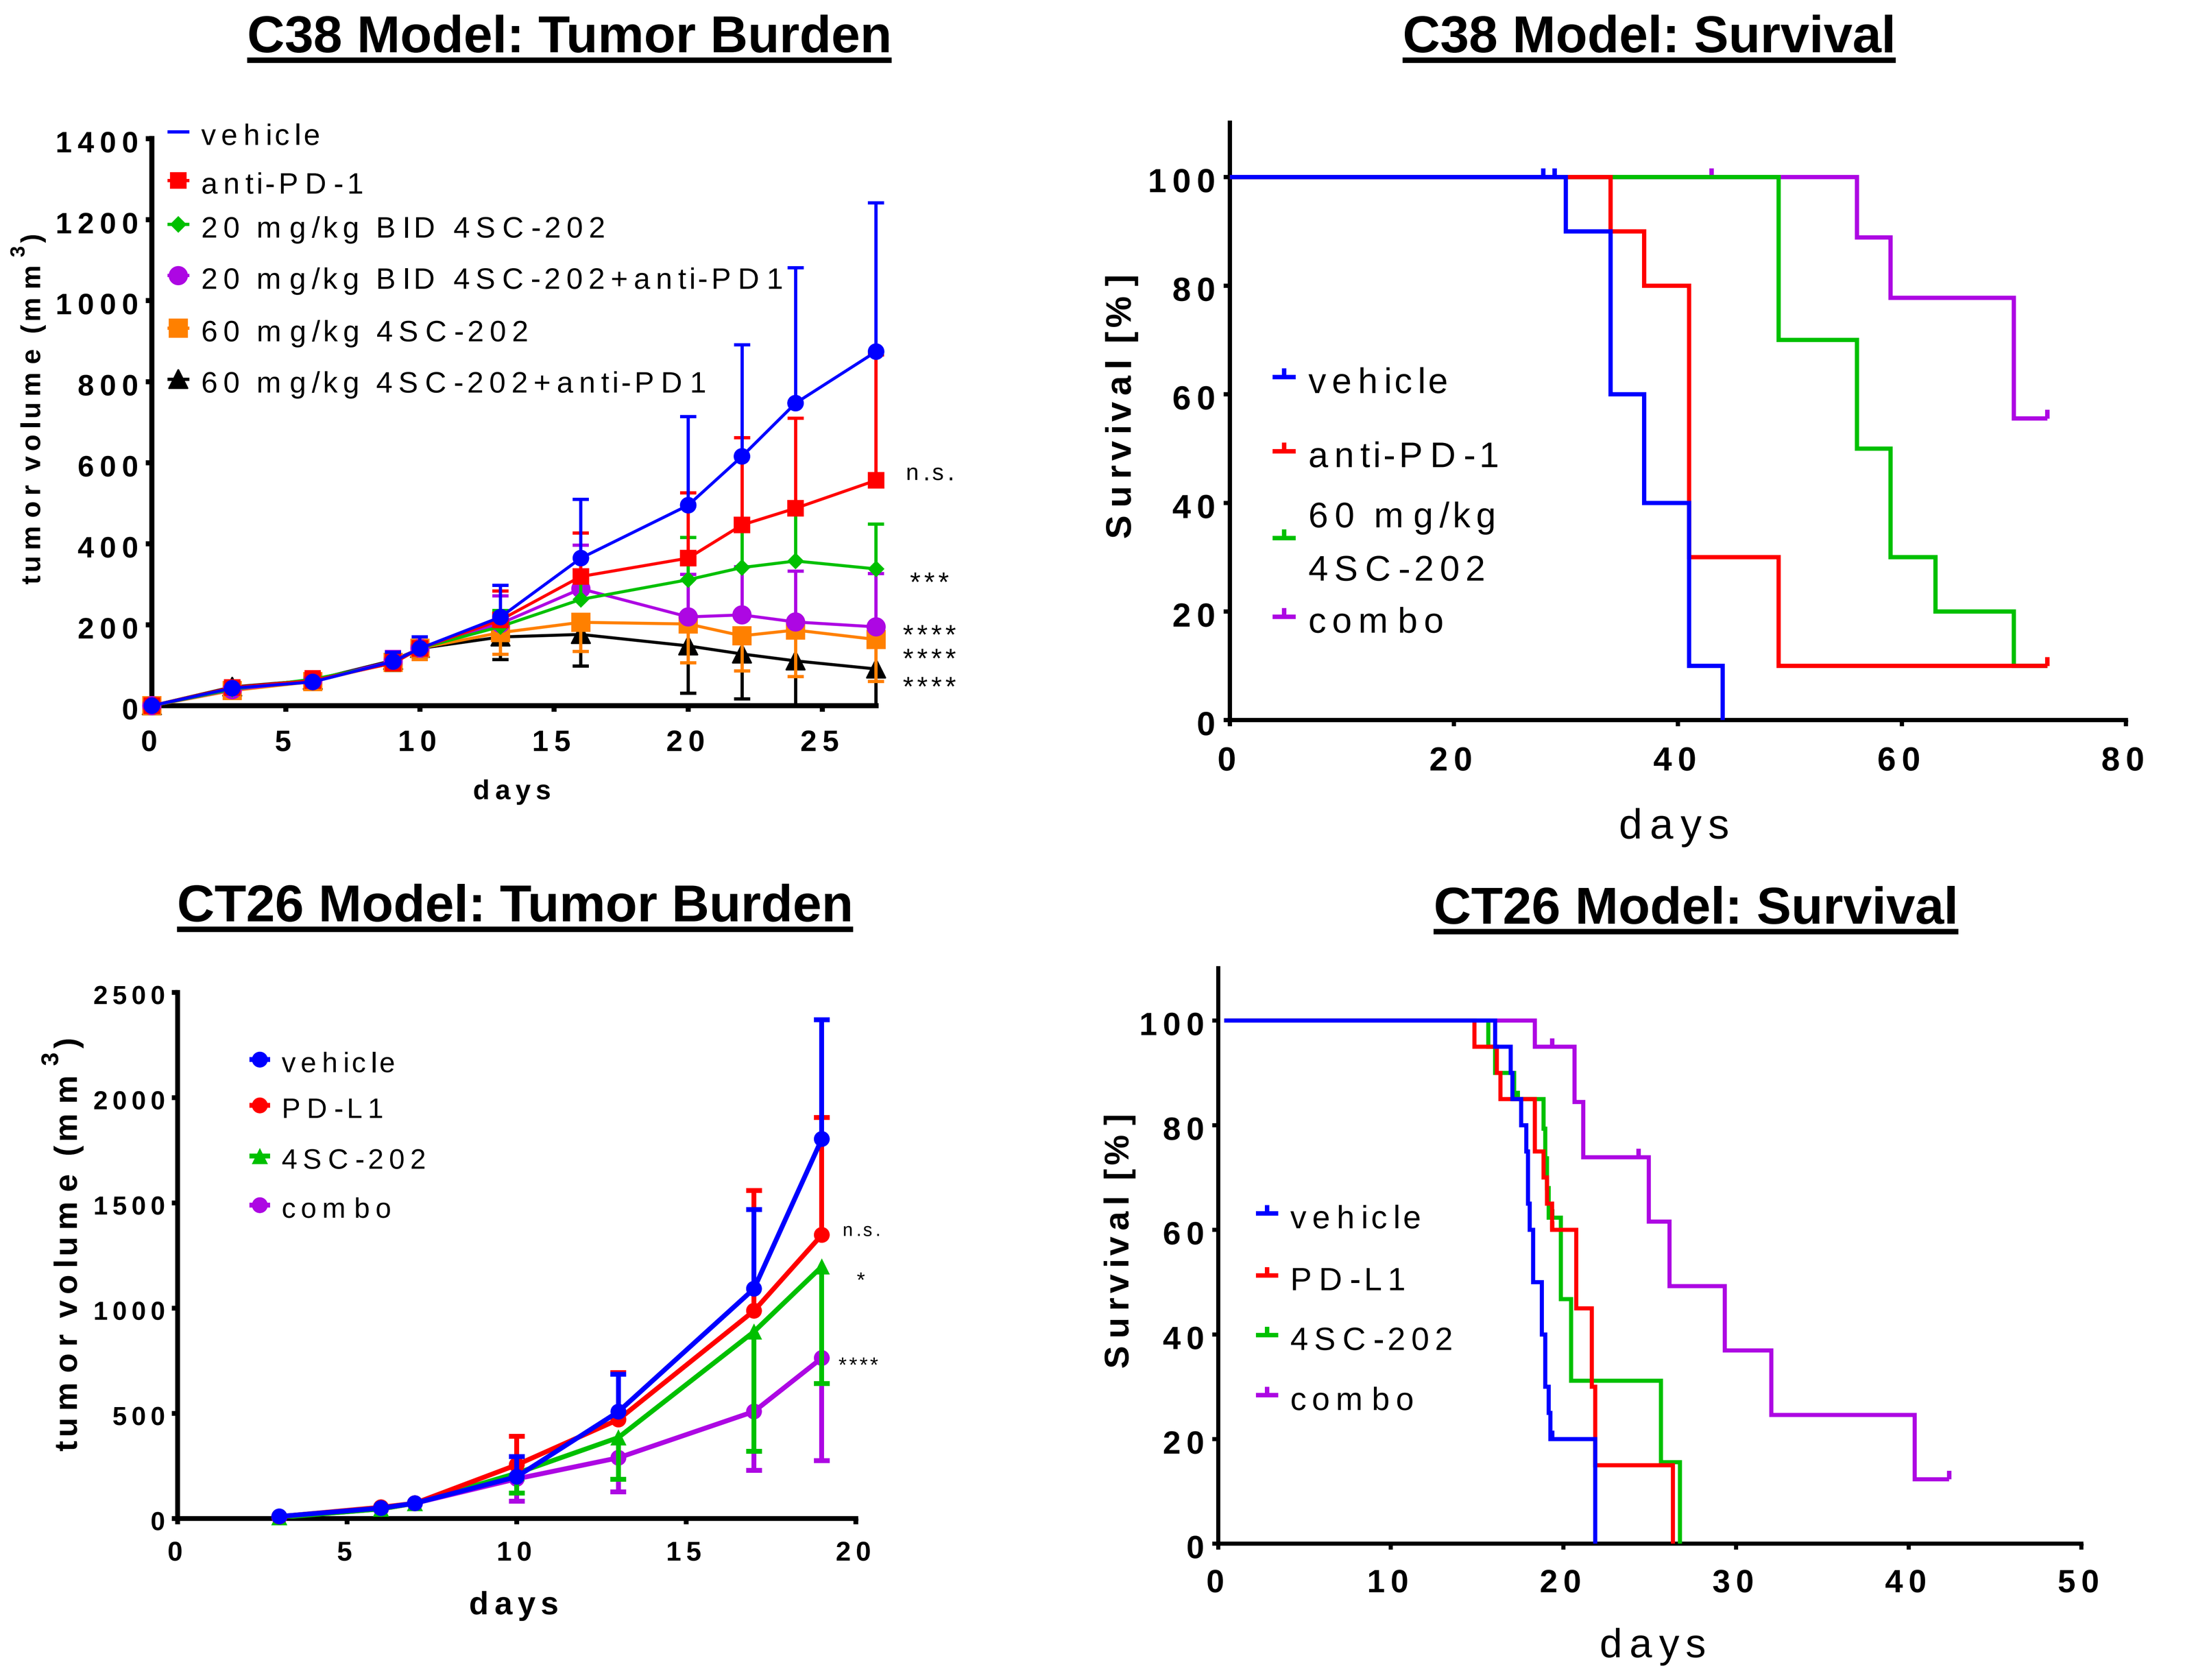

C38 Model: Tumor Burden
vehicle
anti-PD-1
20 mg/kg BID 4SC-202
20 mg/kg BID 4SC-202+anti-PD1
60 mg/kg 4SC-202
60 mg/kg 4SC-202+anti-PD1
tumor volume (mm3)
days
n.s.

C38 Model: Survival
Survival [%]
vehicle
anti-PD-1
60 mg/kg 4SC-202
combo
CT26 Model: Tumor Burden
vehicle
PD-L1
4SC-202
combo
CT26 Model: Survival

Fig. 2: Combination of 4SC-202 and checkpoint inhibitor reduces tumor burden and increases survival in animal models.

SENSITIZE study concept and design

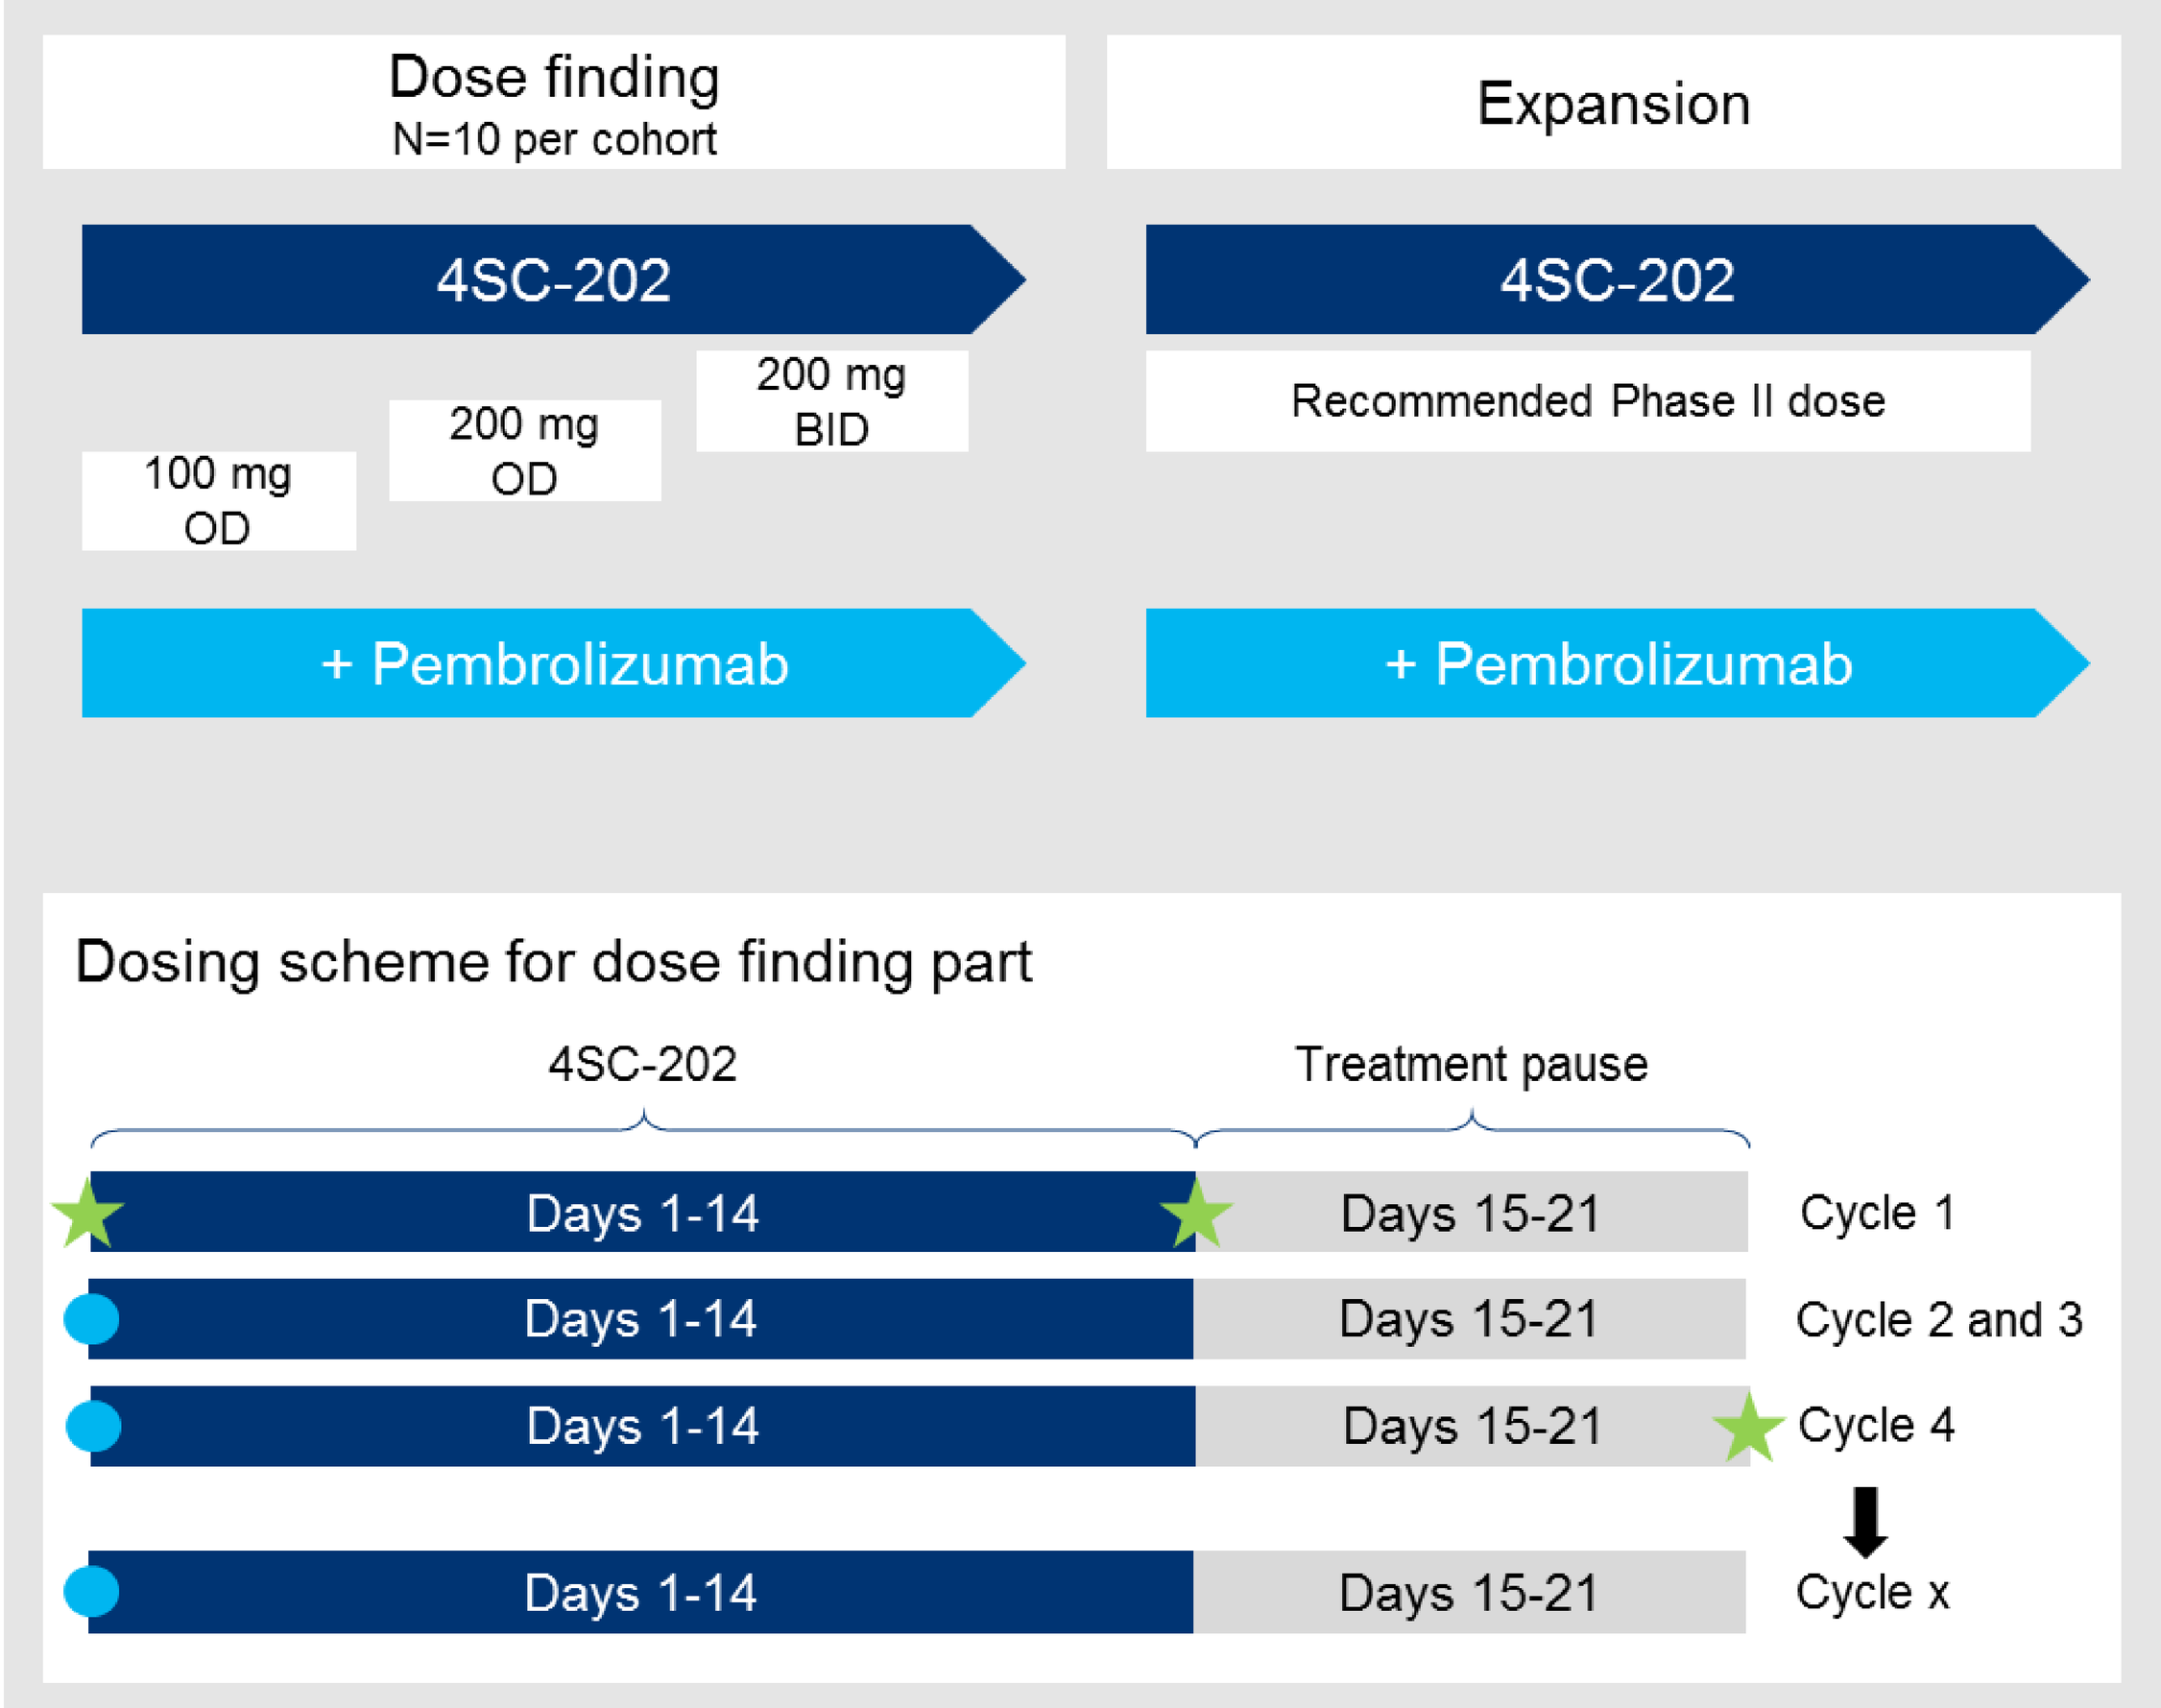

Dose finding
N=10 per cohort
Expansion
4SC-202
100 mg OD
200 mg OD
200 mg BID
Recommended Phase II dose
+ Pembrolizumab
Dosing scheme for dose finding part
4SC-202
Treatment pause
Days 1-14
Days 15-21
Cycle 1
Cycle 2 and 3
Cycle 4
Cycle x

Study Overview:

- Patients with unresectable stage III or stage IV cutaneous melanoma
- Primary refractory or non-responding to anti-PD-1 monotherapy
- n=30 patients in 6 certified skin cancer center in Germany
- 3 dose cohorts [100, 200, 2 x 200 mg 4SC-202 + Pembrolizumab 2 mg/kg q3w]

Primary Endpoint:

- Safety and tolerability

Secondary Endpoints:

- PK/PD Analysis
- Antitumor activity per irRECIST

Exploratory Endpoints:

- Biomarker assessment
- Gene expression tumor and blood
- IHC Analysis (tumor)

Conclusions

Conclusion: Several preclinical tumor models have implicated remarkable immunomodulatory effects of 4SC-202 and the synergistic potential of combining with a checkpoint inhibitor. This epigenetically-driven modulation of the immune and tumor microenvironment by the HDAC class I/LSD1 Inhibitor 4SC-202 in Anti-PD1 refractory/non-responding cutaneous melanoma patients is hypothesized to sensitize the progressive tumor for the synergistic consecutive treatment of 4SC-202 in combination with Pembrolizumab to achieve clinical benefit. The Phase Ib/II SENSITIZE study will test three dose cohorts of 4SC-202 for safety, tolerability and anti-tumor activity. A comprehensive biomarker panel analysis will be performed in sequential biopsies to investigate microenvironment changes on protein, gene expression and exosome level.

The SENSITIZE study is planned to enroll 30 patients in up to 6 certified skin cancer center in Germany and recruitment will start this month. Topline data are expected to be available in 2018.

Contact and funding

René Bartz
Frank Hermann

rene.bartz@4sc.com
frank.hermann@4sc.com

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors."

GEFÖRDERT VOM
Bundesministerium für Bildung und Forschung

The EMTherapy project is conducted in the framework of the European Eurostars program and has received funding from the Federal Ministry of Education and Research.